Sophion Bioscience A/S
Baltorpvej 154, 2750 Ballerup,
Denmark
Phone: +45 4460 8800
info@sophion.com
www.sophion.com

Voltage- and current clamp on induced pluripotent cardiomyocytes with automated patch clamp

Daniel Sauter, Anders Lindqvist, Rasmus Bjørn Jacobsen, Jens Henneke, Mads P.G. Korsgaard

Introduction

Ion channels are large protein molecules imbedded in the membrane of all living cells. The play crucial roles in the electrical and chemical signaling between cells in all organisms. Consequently they constitute good pharmacological targets and have given rise to their own category in pathophysiology, namely channelopathy. This is when ion channel malfunction is the cause of a disease. Additionally, ion channels are involved in the symptomatology of almost all disease areas.

The characterization and modulation of ion channel behavior is most accurately done with electrophysiology. This used to be a laborious process with manual patch clamp but now a days patch clamp has been automated and automatic patch clamp (APC) is playing an ever increasing role in drug discovery. APC has also entered the 384-arena which opens for electrophysiological screening already in the primary phase of drug discovery giving rise to better lead molecules.

Screening is usually done with voltage-clamp techniques. However, current clamp is also relevant when describing more complex ion channel behavior. We here show data from Sophion Qube in 384 format demonstrating how voltage-clamp and current clamp recordings can be carried out interchangeably on the same cell during an experiment. Using cell lines as well as iPS cells we show the benefit of the large number of simultaneous recordings for interpretation of the cellular response.

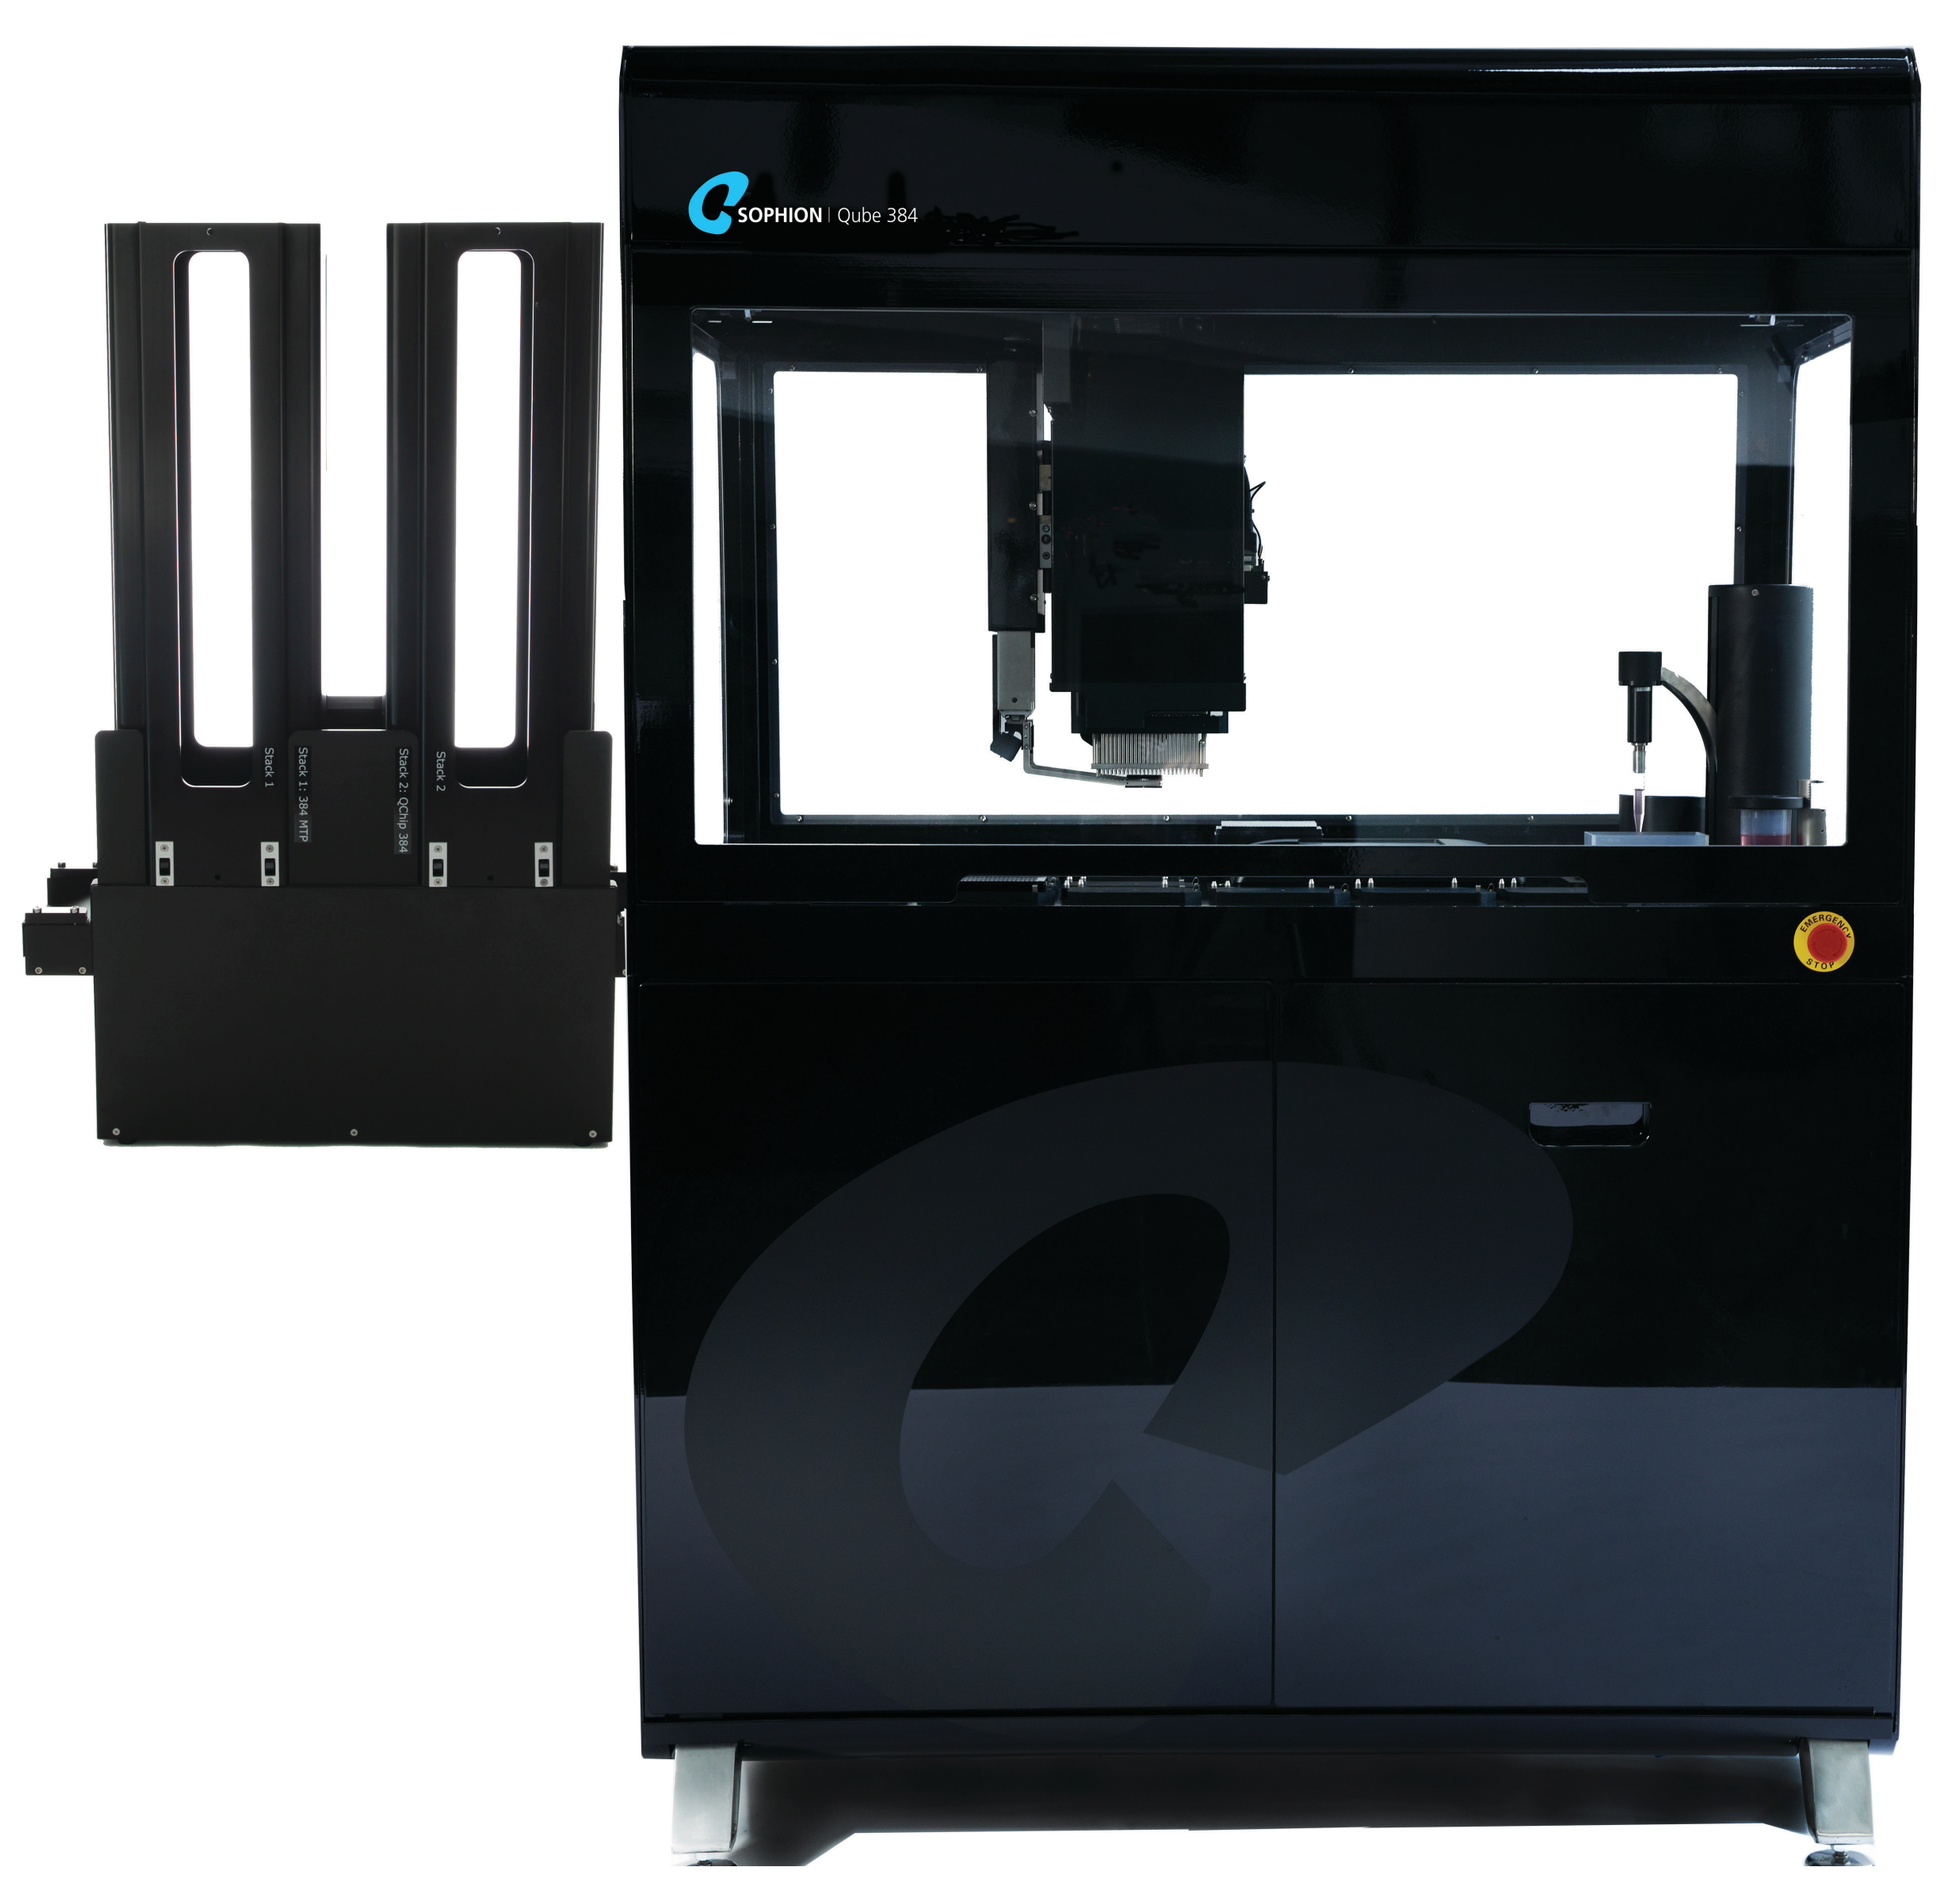
SOPHION | Qube 384

Fig. 1: Qube 384. With 384 parallel measurement sites and 10–15 minutes per plate run, the Qube enables testing of more than 1,500 compounds on different cells in one hour. With full automation features for unattended operation – a plate stacker and a cell preparation unit –, the system provides walk-away functionality for more than 6,000 compounds before user interventions is needed.

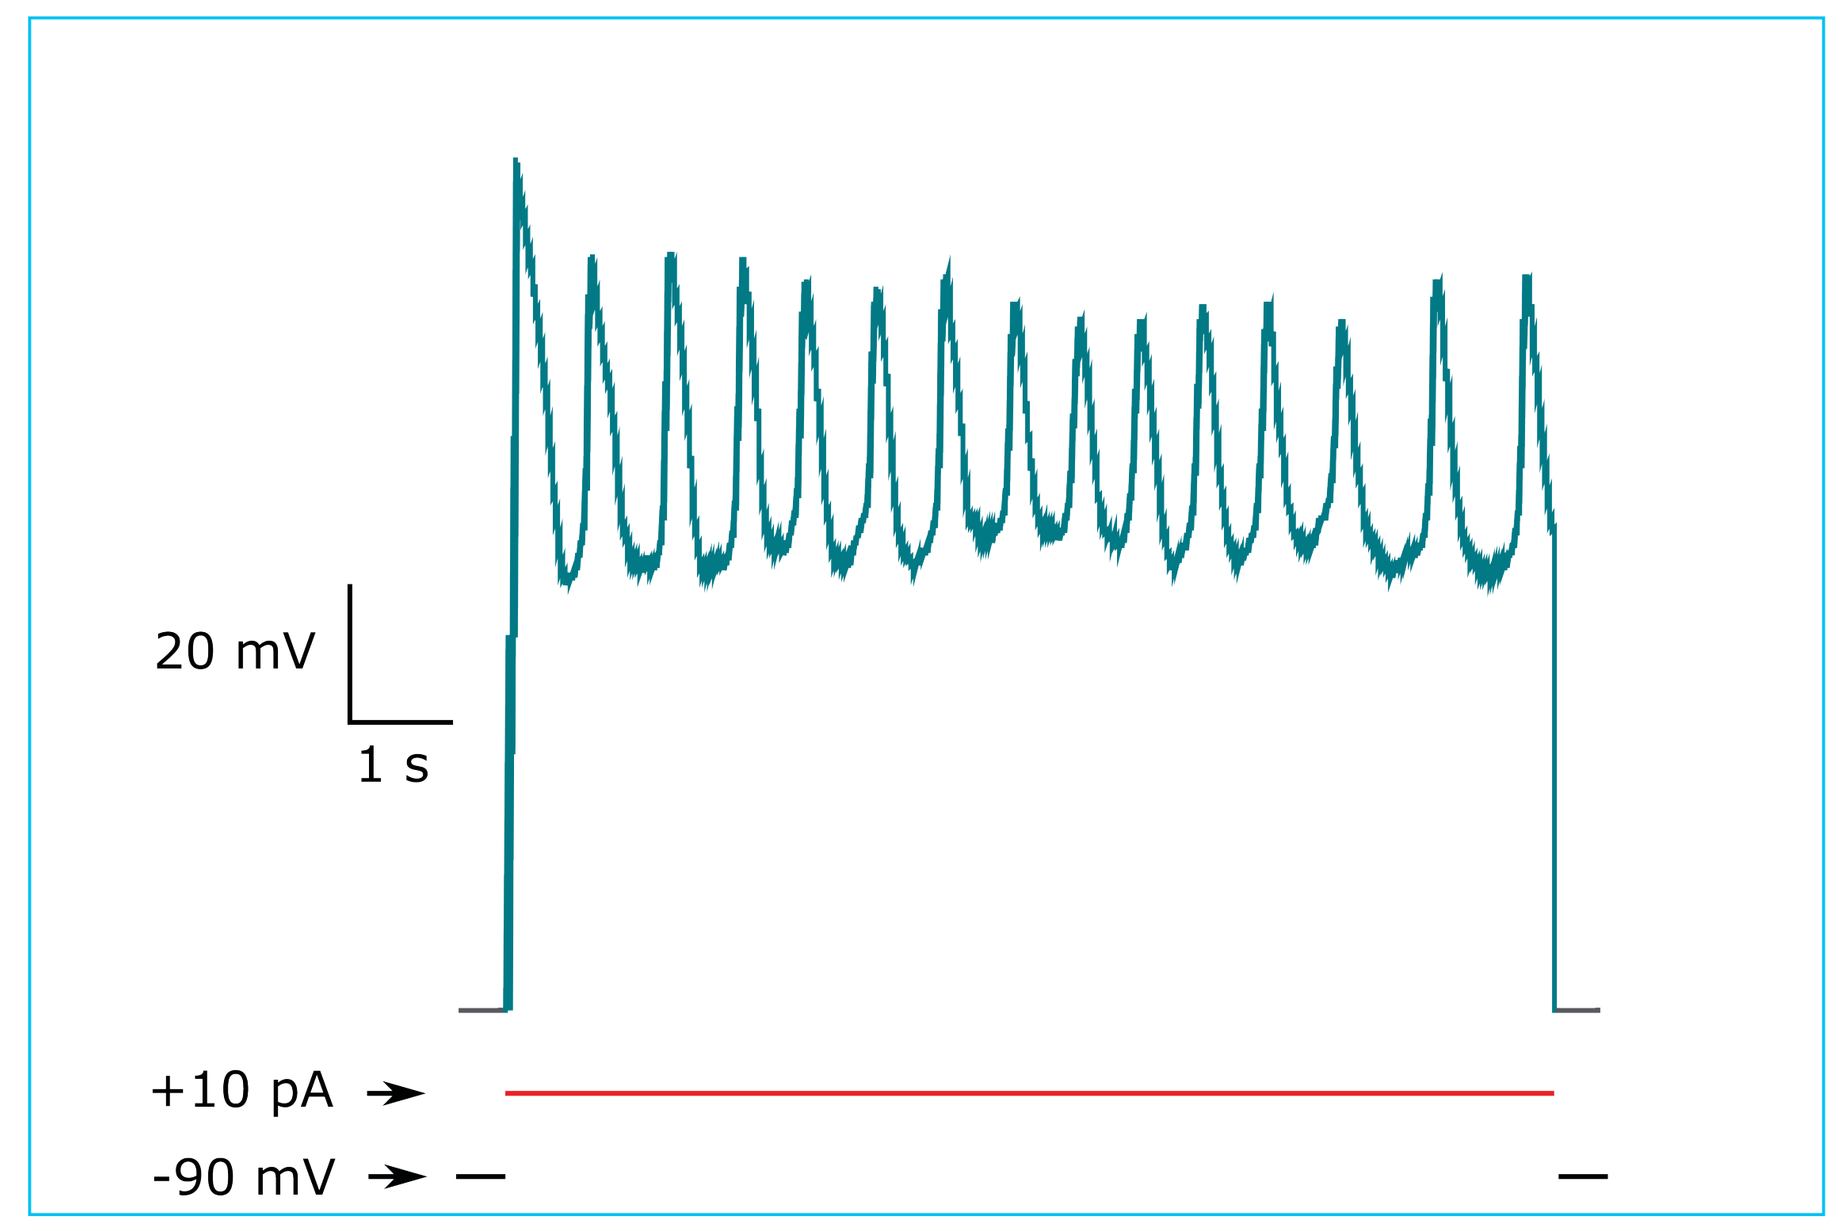
20 mV
1 s
+10 pA
-90 mV

Fig. 2: Using the Qube it is possible to run voltage and current clamp in the same sweep. Action potentials in HL-1 cells induced by current injection. Cells were held at -90 mV (black) and then mode was switched from voltage to current clamp and a +10 pA current was injected (red). At second black line mode was again switched back to voltage clamp.

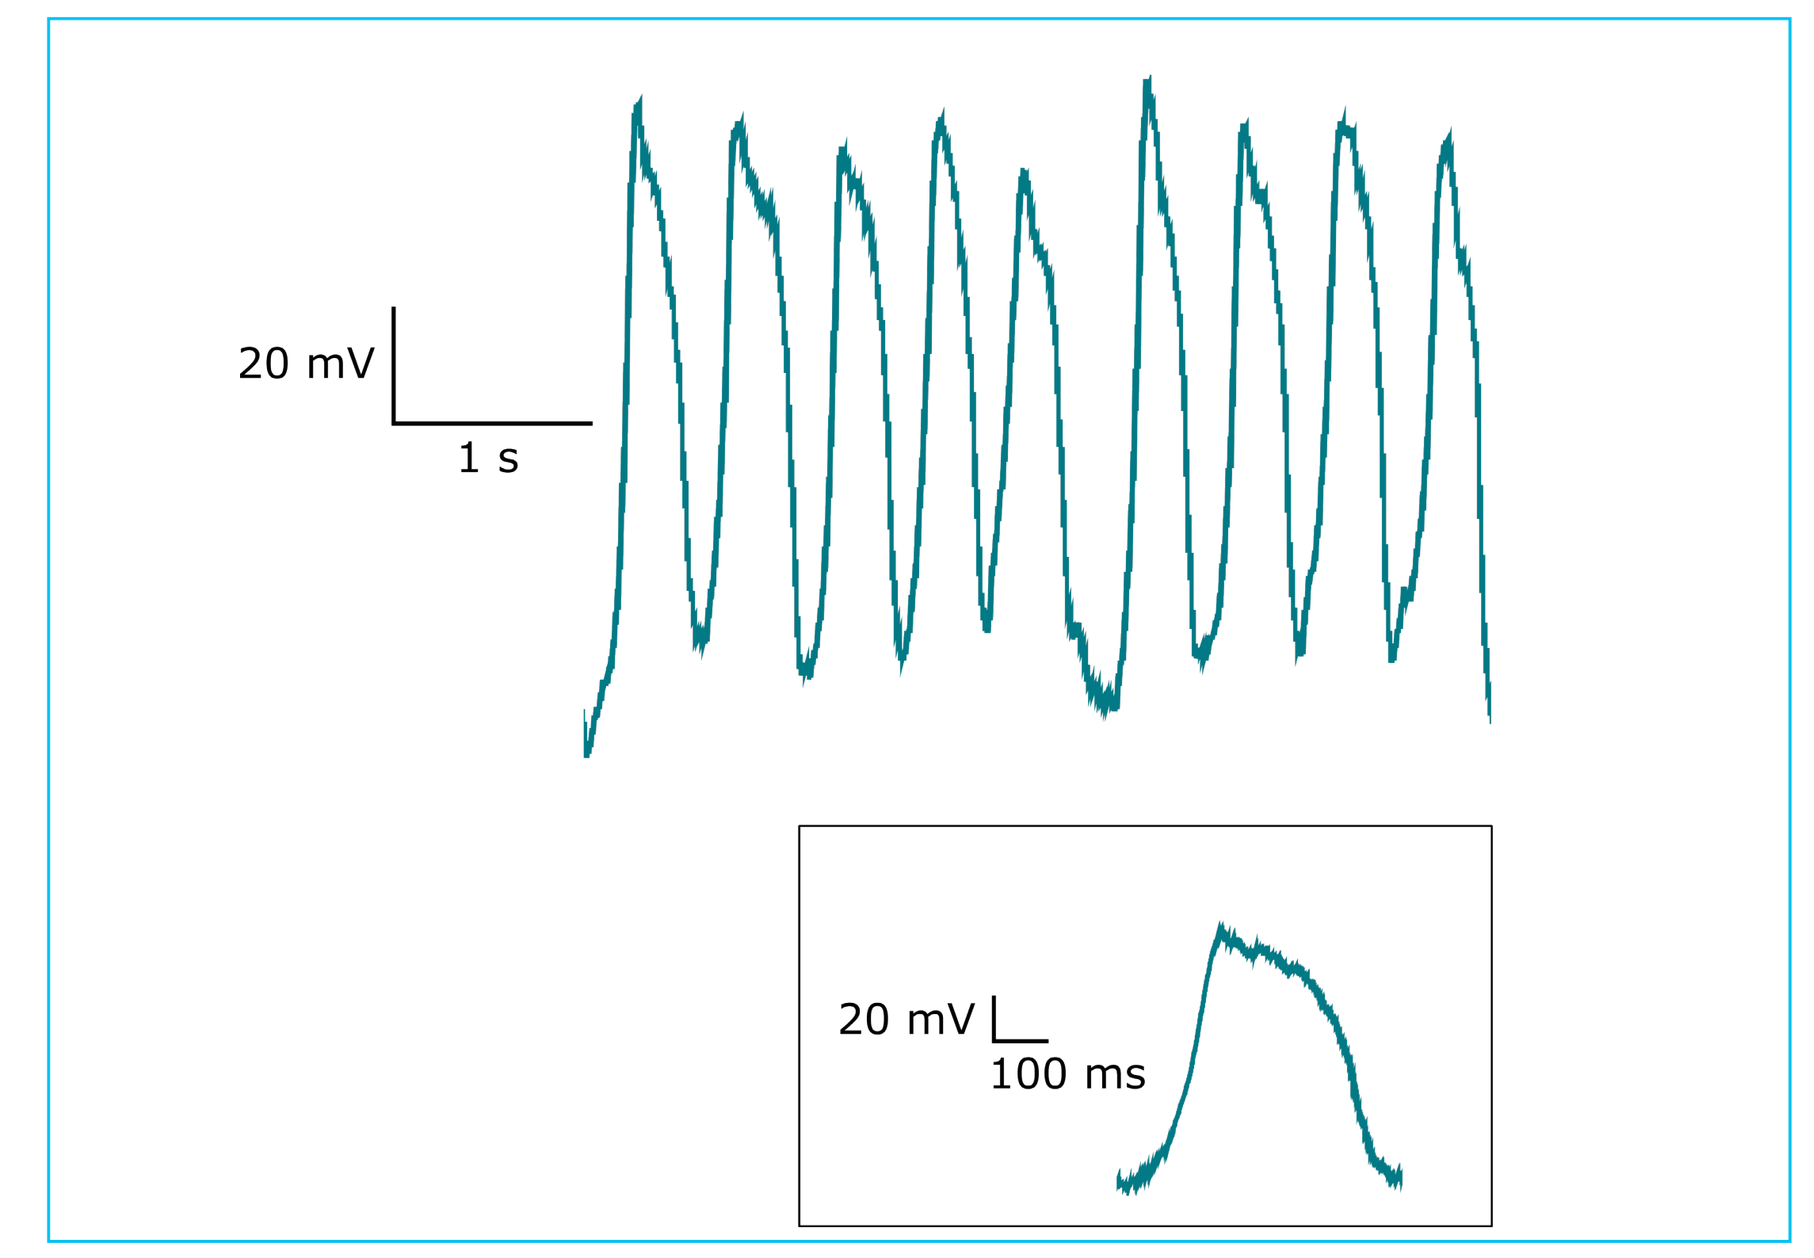
20 mV
1 s
20 mV
100 ms

Fig. 3: Spontaneous action potentials in HL-1 cells recorded in current clamp mode. Insert shows a single action potential on an expanded time scale.

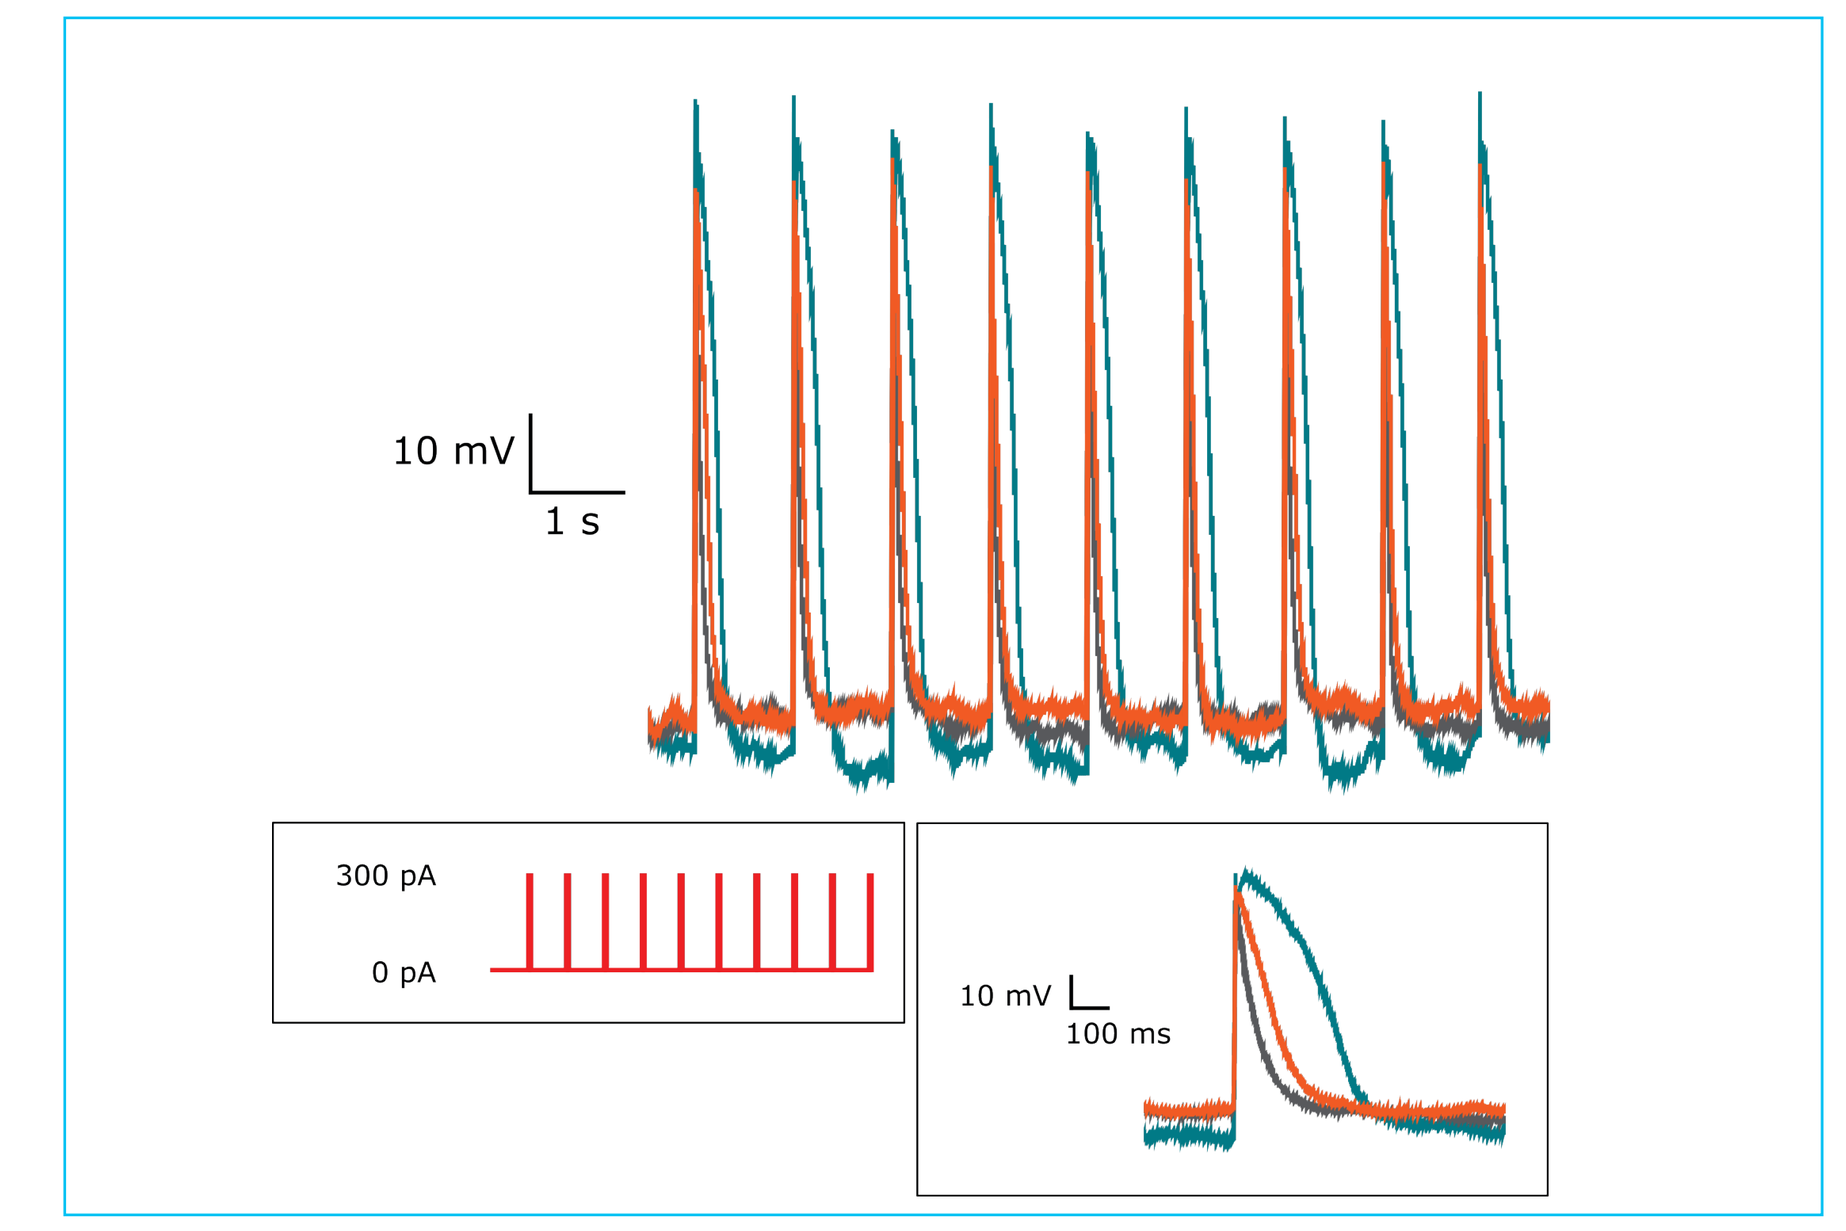
10 mV
1 s
300 pA
0 pA
10 mV
100 ms

Fig. 4: Induced action potentials in HL-1 cells. Cells were held at 0 pA holding current and paced at 1 Hz using 300 pA current injections (3 ms duration) under control conditions (green trace) and in the presence of 100 nM (orange) and 1 µM verapamil (grey). Inserts show a single action potential on an expanded timescale and current protocol.

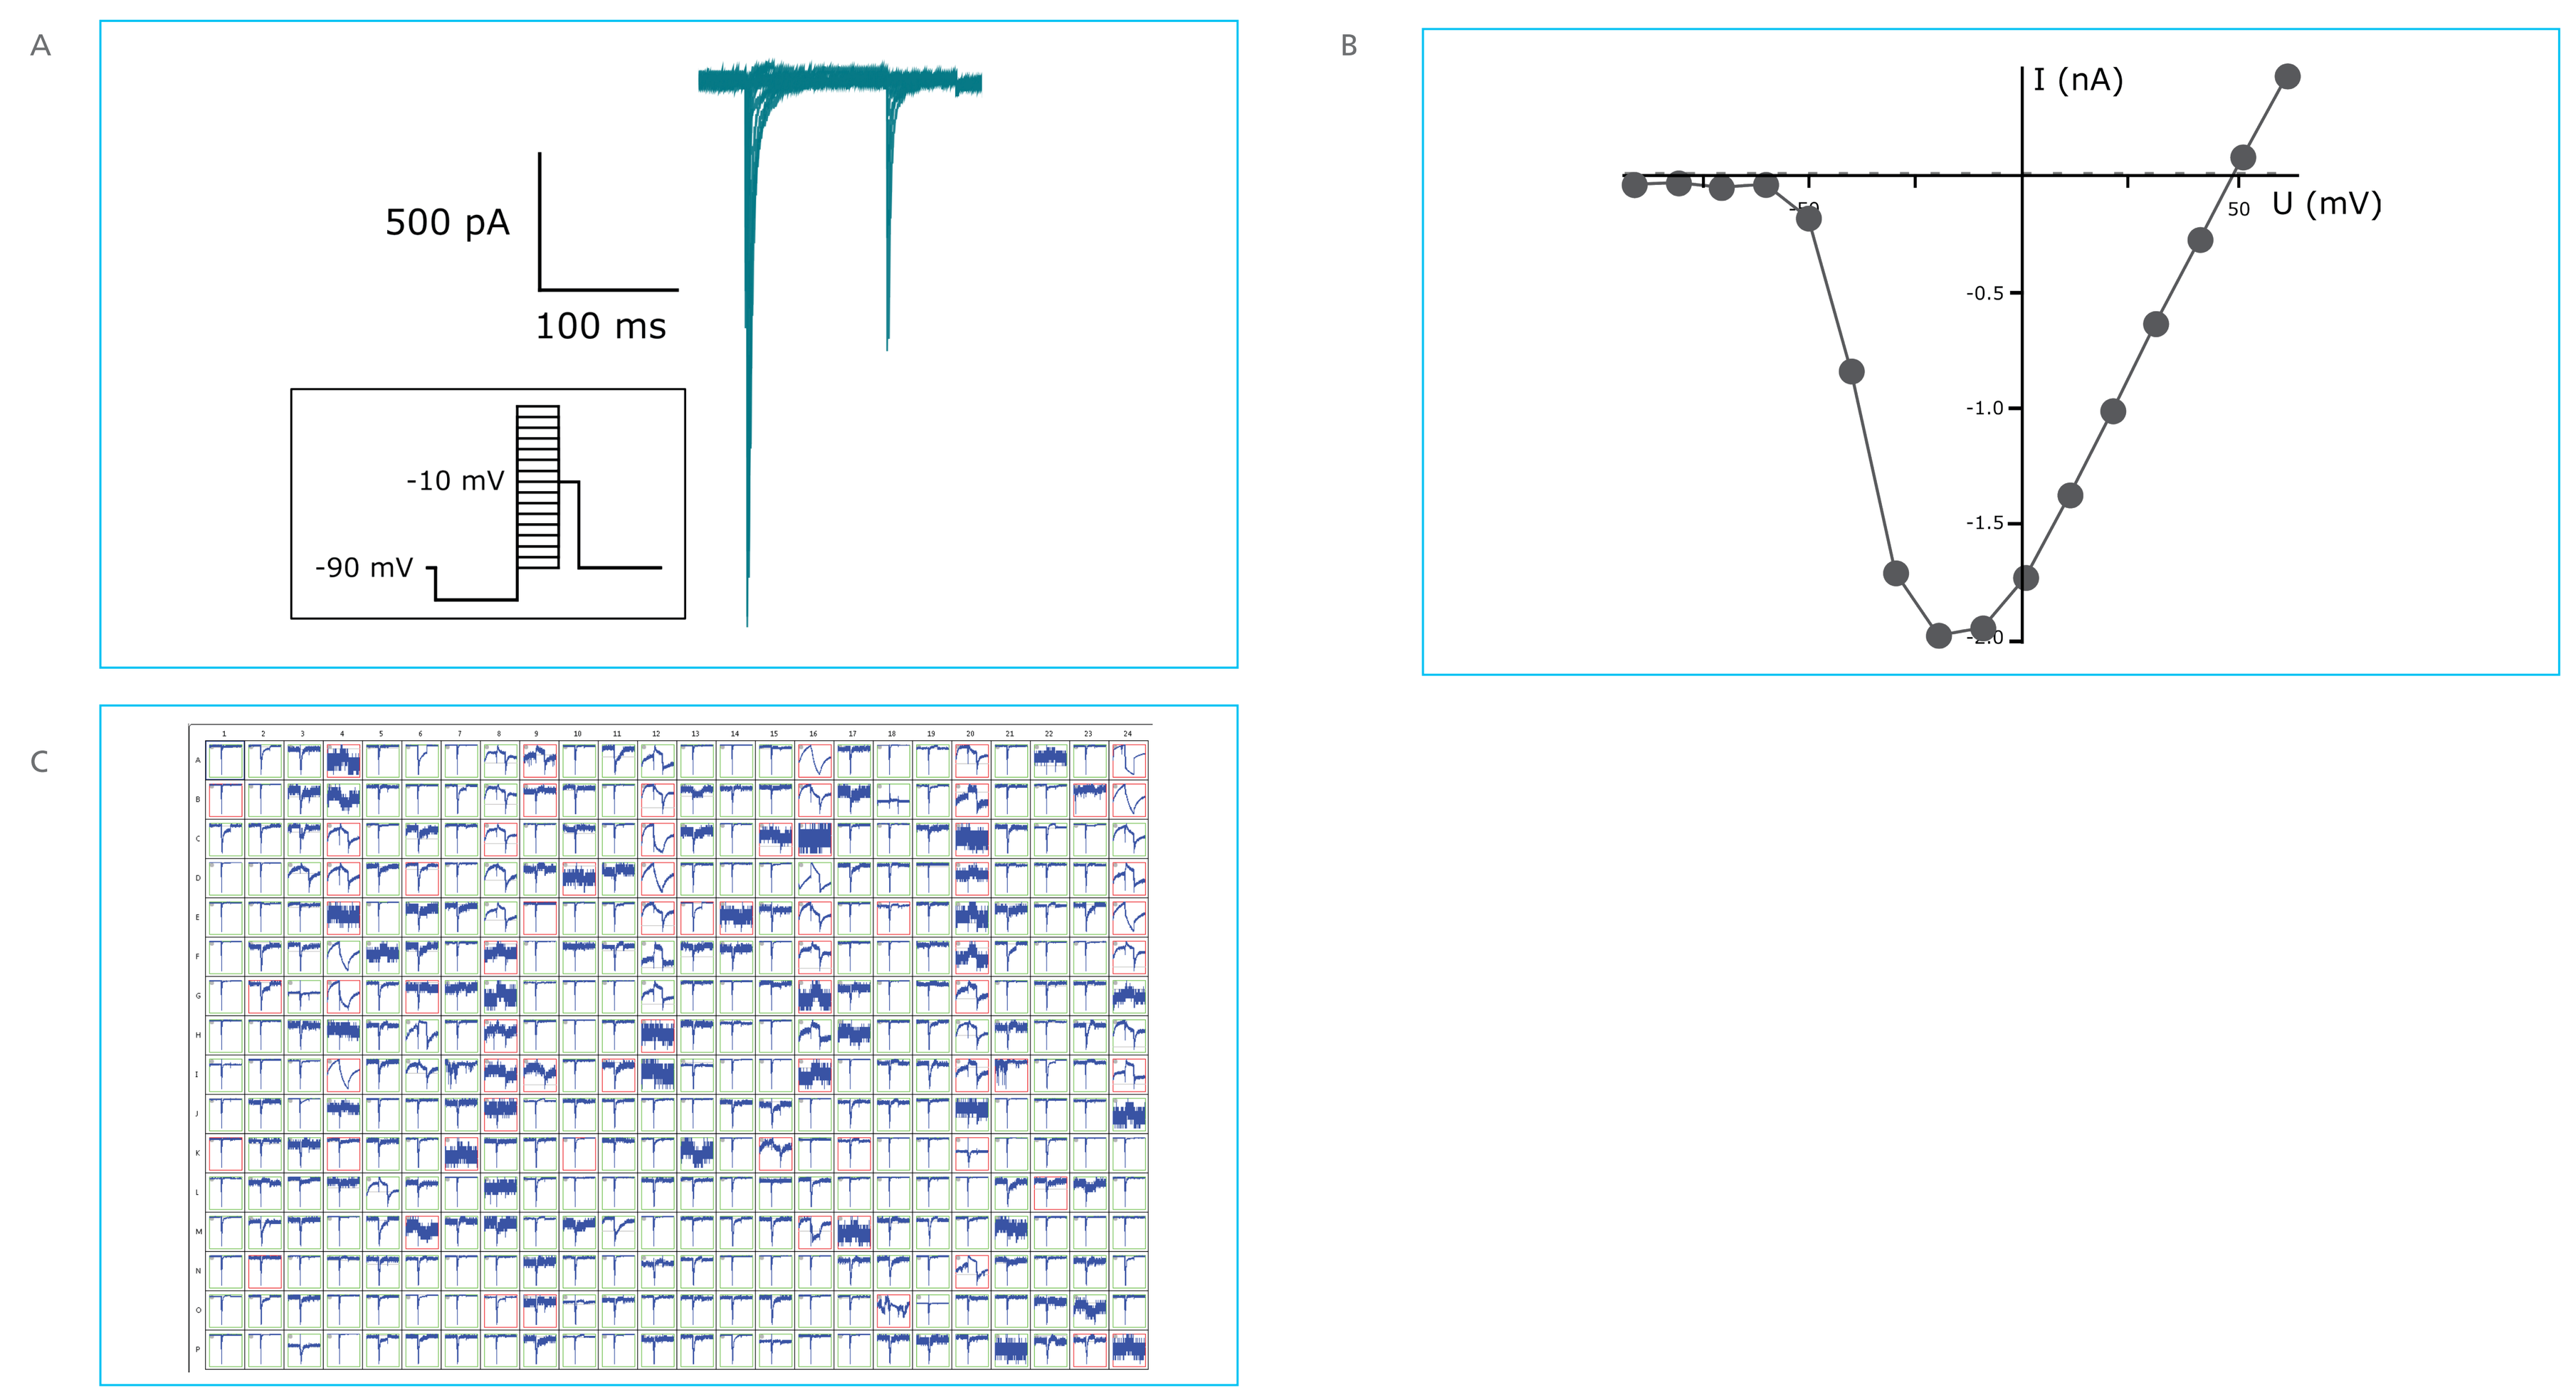
A
500 pA
100 ms
-10 mV
-90 mV
B
I (nA)
U (mV)
50
-0.5
-1.0
-1.5
C

Fig. 5: Current-voltage relationship for sodium currents in HL-1 cells. A) Representative current traces evoked by depolarizing from -120 mV to different test potentials in steps of 10 mV. Insert shows voltage protocol. B) Current-voltage relationship and C) plate view of QChip.

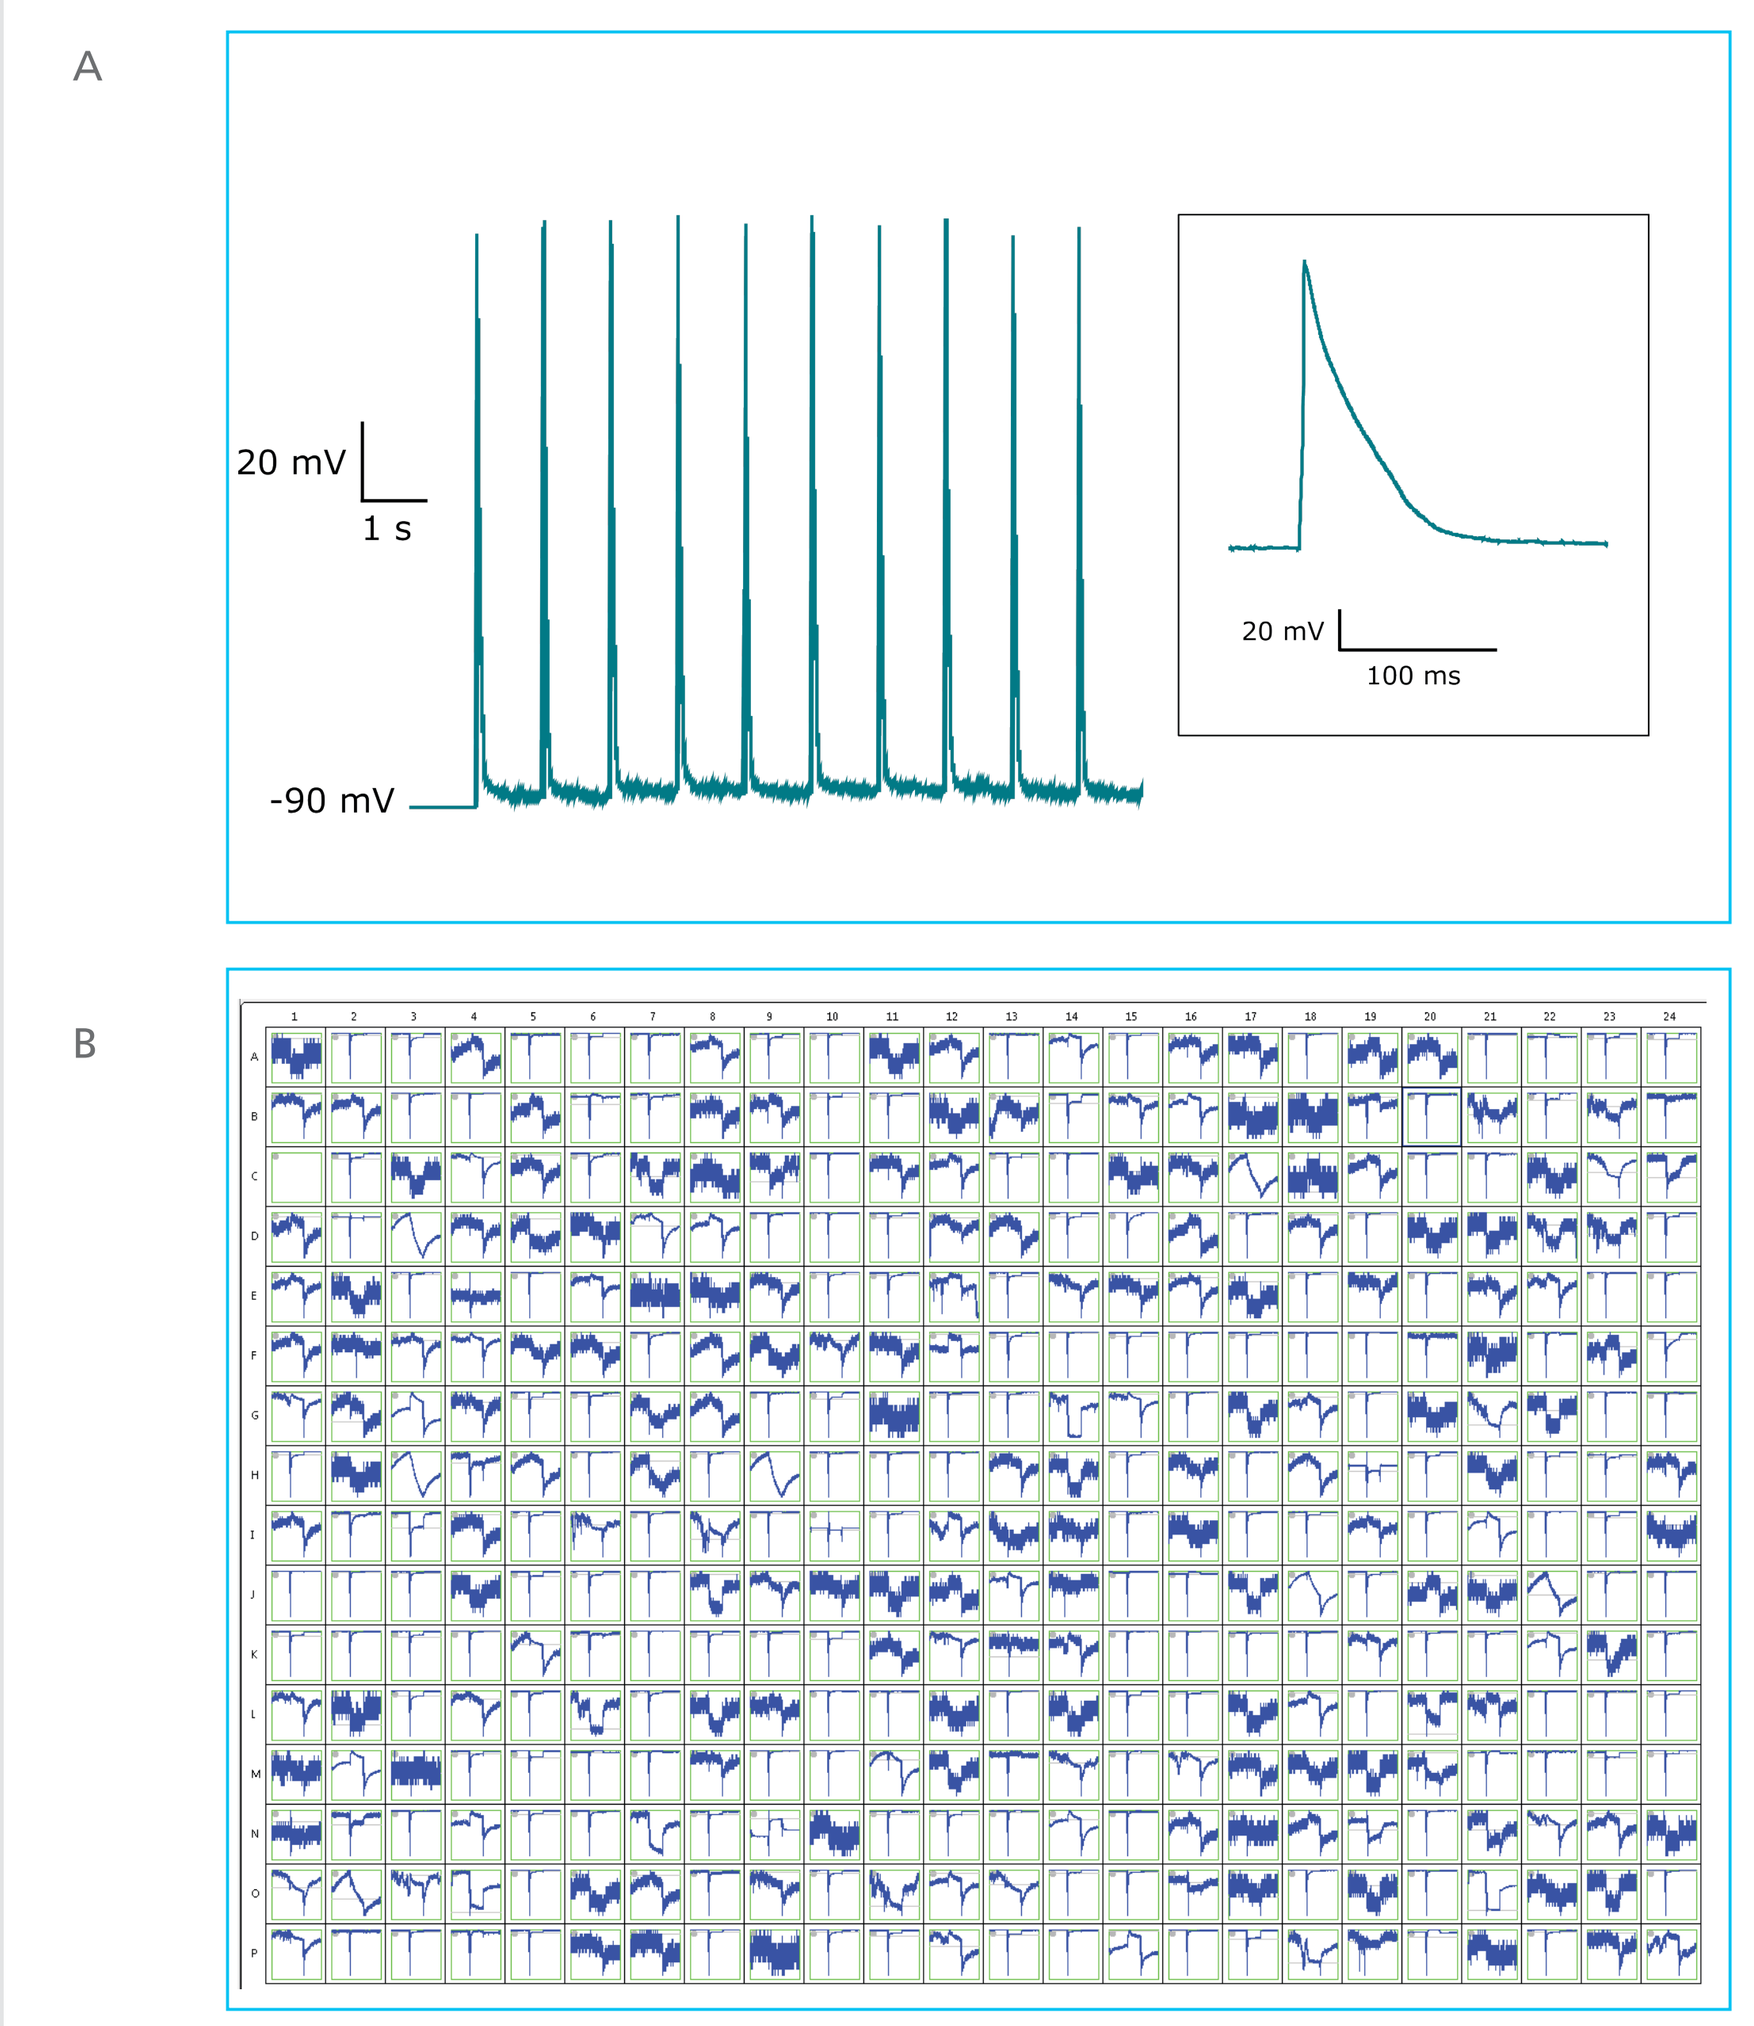
A
20 mV
1 s
-90 mV
20 mV
100 ms
B

Fig. 6: Action potentials in Axiogenesis© COR.4U human iPS cell-derived cardiomyocytes A) Action potentials induced by current injection pulses (500 pA, 3 ms, 1 Hz). Insert shows a single action potential on an expanded time scale. The holding current between pulses was 0 pA. B) Plate view showing inward currents. Voltage protocol identical to the one shown in Fig. 5.

Conclusion

Here we show experiments in voltage and current clamp modes on Qube 384 using HL-1 mouse atrial cardiomyocytes and Axiogenesis© matured COR.4U iPS cell-derived cardiomyocytes. In both cell types it was possible to evoke action potentials by injecting current pulses whereas HL-1 cells also showed spontaneous action potentials. In both HL-1 and iPS CMs it was possible to evoke inward sodium currents and in a fraction of the cells also calcium currents (not shown). A distinguished feature of the Qube is the possibility to combine both current and voltage clamp recordings in the same sweep making it possible to carry out very fine-tuned experiments.

References

Claycomb WC, Lanson NA Jr, Stallworth BS, Egeland DB, Delcarpio JB, Bahinski A, Izzo NJ Jr. "HL-1 cells: a cardiac muscle cell line that contracts and retains phenotypic characteristics of the adult cardiomyocyte". Proc Natl Acad Sci U S A. 95:2979-84, 1998.

Materials and methods

HL-1 cells were cultured as described in Claycomb et al. (1998). iPS-derived cardiomyocytes (CMs) were kindly provided by Axiogenesis AG (Cologne, Germany) and cultured according to instructions provided by Axiogenesis. Experiments with iPS-derived CMs were carried out at 20 degrees and experiments with HL-1 at ambient temperature.

Presented @Biophysics2017